A Novel Aseptically Processed Human Reticular Dermis* Promotes Fibroblast Secretion of Growth Factors that Support Angiogenesis and Epithelialization in Wound Healing;

[1]Dasgupta, A; [1]Huang, YC; [1]Imming, E; [1]Gidwani, M; [1]Eng S; [1]Chnari, E

[1]Musculoskeletal Transplant Foundation (MTF), Edison, NJ, USA

INTRODUCTION

Dermal scaffolds provide a natural architecture for tissue replacement in acute and chronic wounds. The inherent extracellular matrix supplies both structural and signaling cues for host cell infiltration, integration and long-term remodeling. Upon cell integration, the dermal scaffold can retain cell secreted matrix proteins and growth factors to modulate host microenvironment interactions and support the wound healing cascade. Traditional acellular dermal matrices (ADMs) are non-uniformly porous leading to asymmetric cell infiltration which may prolong host cell integration. We have previously shown that aseptically processed reticular dermis is uniformly porous promoting faster, symmetrical fibroblast infiltration resulting in an abundance of secreted matrix proteins; these are important attributes in granulation leading to wound closure.

The goal of this study was to further characterize the role of human reticular acellular dermal matrices (HR-ADMs) in supporting cell secreted growth factors that would promote angiogenesis. The impact of these released trophic factors on *in vitro* angiogenic activities of endothelial cells were investigated as a single culture and in co-cultures with fibroblasts.

MATERIALS AND METHODS

HR-ADMs were procured according to Good Tissue Practices and processed aseptically without terminal sterilization at Musculoskeletal Transplant Foundation (MTF, Edison NJ, USA).

Normal human dermal fibroblasts (NHDFs) were cultured (0.3 million cells/7mm disc) on pre-hydrated HR-ADMs and the conditioned media was removed over time *(Figure 1A)*. Quantitative microarray analysis (RayBioTech) was conducted to elucidate growth factors secreted by cultured fibroblasts. This conditioned media was used to examine *in vitro* angiogenic capacity of human umbilical vascular endothelial cells (HUVECs) through tubular formation (Cultrex). Additionally, co-cultures of NHDFs and HUVECs (2:1 ratio) were mixed at day 0 *(Figure 1B)* and sequentially cultured *(Figure 1C)* to examine tubular formation (CD31), secretion of angiogenic factor, von Willebrand Factor (vWF) and matrix protein secretion (laminin, collagen IV, fibronectin) over time through confocal (Rutgers University) and histological imaging (IHC World, LLC).

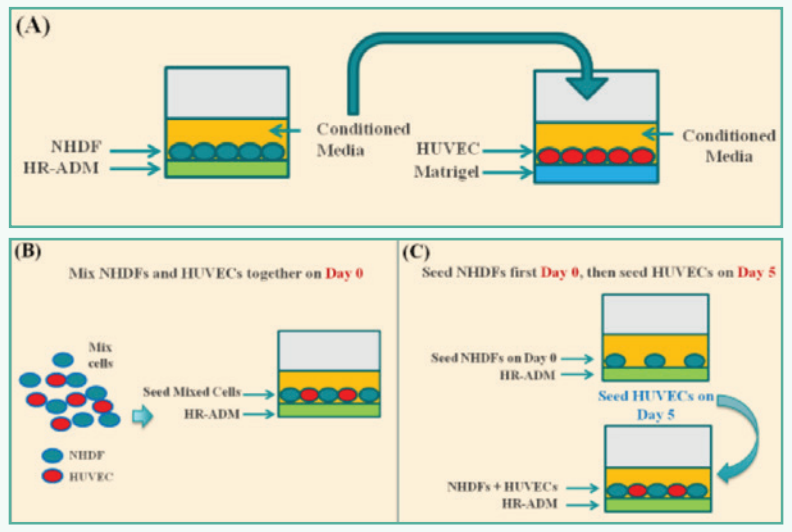
(A)
NHDF
HR-ADM
Conditioned Media
HUVEC
Matrigel
Conditioned Media
(B)
Mix NHDFs and HUVECs together on Day 0
Mix cells
Seed Mixed Cells
HR-ADM
NHDF
HUVEC
(C)
Seed NHDFs first Day 0, then seed HUVECs on Day 5
Seed NHDFs on Day 0
HR-ADM
Seed HUVECs on Day 5
NHDFs + HUVECs
HR-ADM

Figure 1: A) NHDF conditioned media was evaluated for growth factors and then to culture HUVECs to examine angiogenic capacity. Co-culture of NHDFs and HUVECs: B) Cells mixed at Day 0 and cultured until Day 10. C) Sequential culture of NHDFs first at Day 0, with HUVECs added at Day 5 and cultured until Day 12. Both angiogenesis and matrix synthesis were examined.

Adhered Fibroblasts are Functional & Secreted Their Own Growth Factors

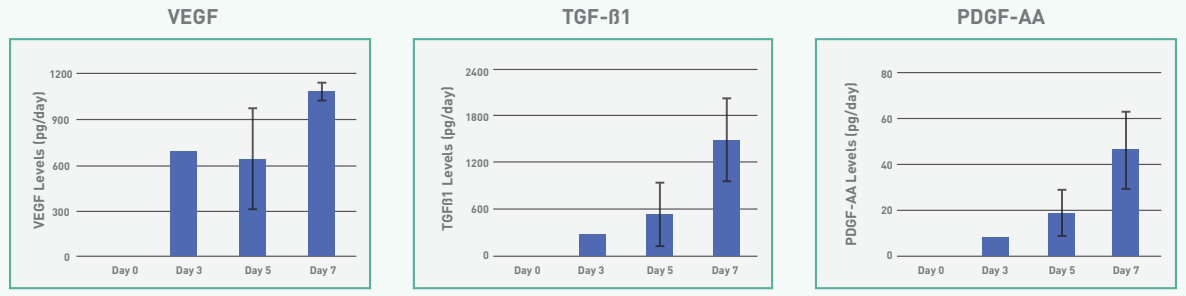
VEGF
VEGF Levels (pg/day)
1200
900
600
300
0
Day 0
Day 3
Day 5
Day 7
TGF-ß1
TGFß1 Levels (pg/day)
2400
1800
1200
600
0
Day 0
Day 3
Day 5
Day 7
PDGF-AA
PDGF-AA Levels (pg/day)
80
60
40
20
0
Day 0
Day 3
Day 5
Day 7

Figure 2: NHDFs cultured on HR-ADMs are functional and started secreting their own growth factors (VEGF, TGF-ß1 and PDGF-AA) as early as day 3 with levels increasing over time. These growth factors are known to support granulation, angiogenic & epithelialization activities during wound healing [1, 2].

Fibroblast Secreted Growth Factors Stimulated Angiogenesis in Endothelial Cells

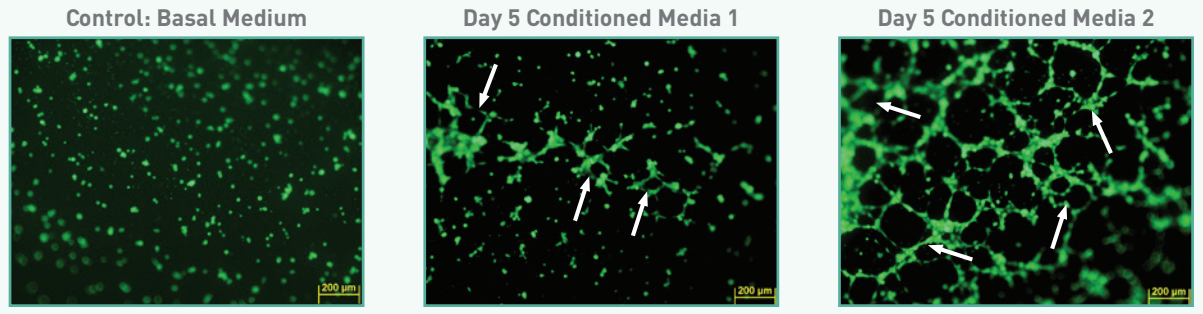
Control: Basal Medium
Day 5 Conditioned Media 1
Day 5 Conditioned Media 2
200 µm
200 µm
200 µm

Figure 3: Conditioned media from cultured NHDFs on HR-ADMs stimulate angiogenesis in HUVECs cultured on Matrigel (magnification 10x). Distinct tubular, network formation (white arrows) was observed at 6 hours compared to the negative control. This highlights functional fibroblast paracrine signaling (VEGF, PDGF-AA), enhancing angiogenic capacity which is an instrumental function during wound healing [1, 2].

Fibroblasts & Endothelial Cells Co-Cultured Together on HR-ADMs Demonstrated Angiogenic Capacity by Day 6

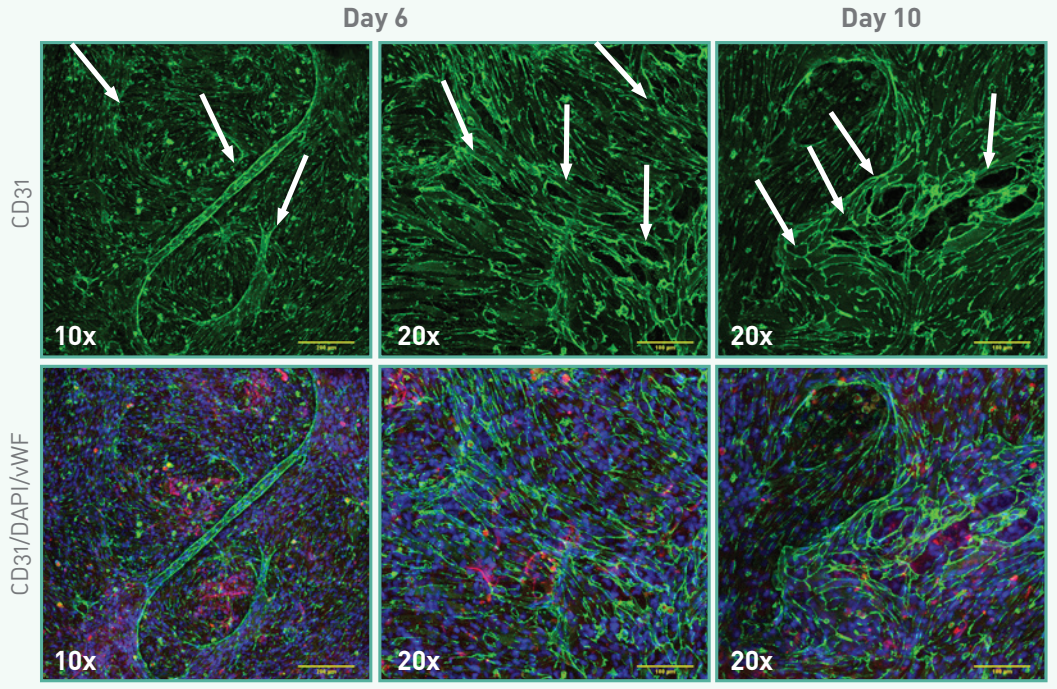
Day 6
Day 10
CD31
CD31/DAPI/vWF
10x
20x
20x
10x
20x
20x

Figure 4: Confocal imaging of co-cultured NHDFs and HUVECs mixed at day 0 were cultured for 6 and 10 days on the deep dermal facing side of HR-ADMs. Cells readily attached as shown by DAPI staining (blue nuclei). Endothelial cell marker, CD31 (green), revealed distinct, sustained tubular, network formation (white arrows), whereas vWF (red) which is secreted by functional endothelial cells also verified angiogenic capacity (10X and 20X magnification). Similar observations were found on both sides of the HR-ADM. This demonstrated the importance of the porous, 3D nature of HR-ADMs along with the retained organized matrix proteins in sustaining angiogenesis over 6 and 10 days in vitro. In contrast, on Matrigel (2-D presentation of basement membrane proteins), the tubular formation is not sustained beyond 24 hours.

Sequential Co-culture of Fibroblasts (Day 0) with Endothelial Cells (Day 5) Showcased Angiogenic Capacity at Day 12

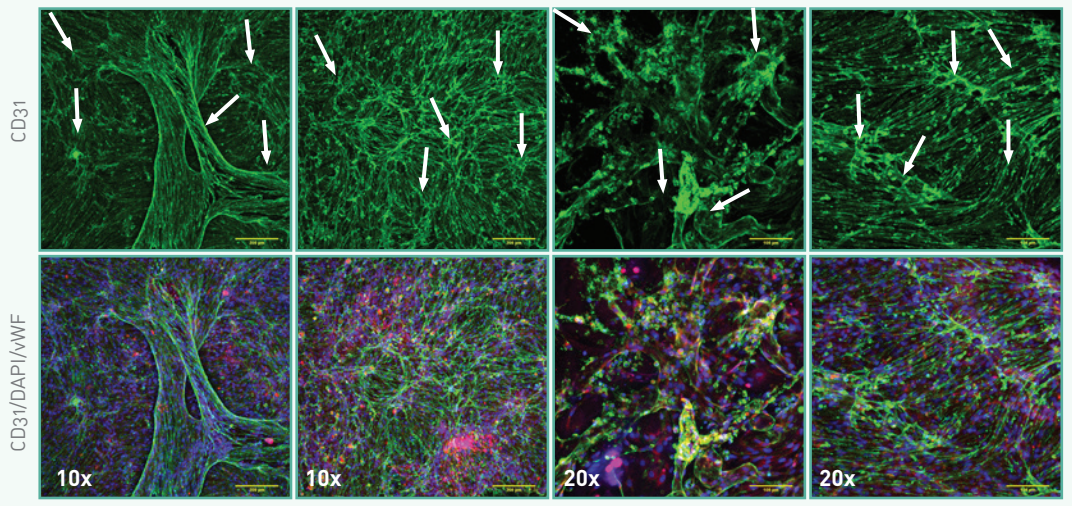
CD31
CD31/DAPI/vWF
10x
10x
20x
20x

Figure 5: Confocal imaging of sequential cultured NHDFs and HUVECs. NHDFs were seeded first (day 0) with HUVECs added at day 5, and cultured until day 12 on HR-ADMs (magnification 10x and 20x). Cells readily attached as shown by DAPI staining (blue nuclei). Angiogenic markers, CD31 (green) and vWF (red), highlight functional endothelial cells, along with distinct tubular, network formation (white arrows) which is characteristic of angiogenesis. Both co-cultured methods yielded extensive angiogenesis by day 12 and illustrate that HR-ADMs can support different stages of wound healing.

Functional Fibroblasts & Endothelial Cells Secrete Matrix Proteins

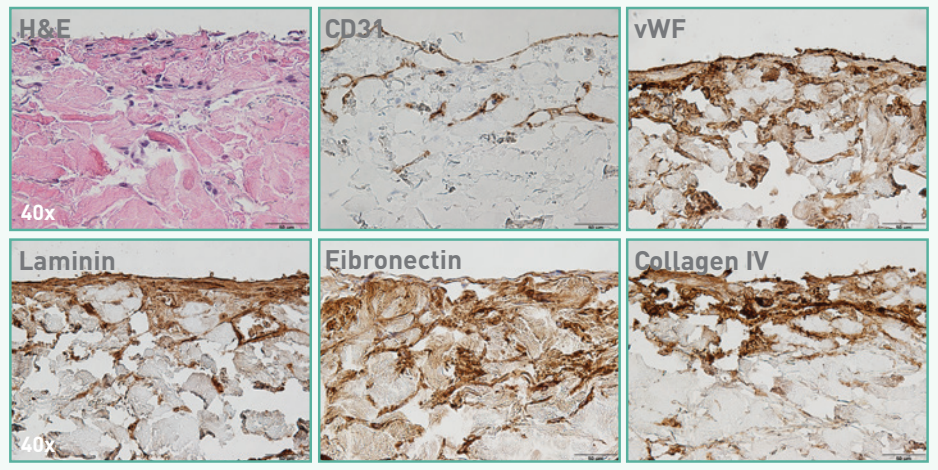
H&E
CD31
vWF
40x
Laminin
Fibronectin
Collagen IV
40x

Figure 6: Histological imaging revealed cellular infiltration (H&E) into the HR-ADM along with extensive staining of endothelial markers (CD31, vWF) surrounding tubular formation of functional HUVECs. In addition, an abundance of matrix production (laminin, fibronectin and collagen IV) can be seen. This positive staining highlights that fibroblast-endothelial co-cultures foster sustained angiogenic and epithelialization activities within HR-ADMs. These are critical steps in the wound healing process [1, 2].

CONCLUSION

The data demonstrates that HR-ADMs provide an ideal scaffold to support cellular activities. Fibroblasts secreted growth factors supported paracrine signaling activities in endothelial cells. In addition, direct co-cultures culminated in extensive angiogenesis and epithelialization within the HR-ADM. This cellular cross-talk is important in supporting wound healing activities.

REFERENCES

1. Vulnar T, *et al. J Int Med Res.* 2009; 2. Reinke JM & Sorg H, *Eur Surg Res.* 2012

©2016 Musculoskeletal Transplant Foundation 4/16 Mtkg -1160 Rev. 0

*AlloPatch® Pliable is a registered trademark of the Musculoskeletal Transplant Foundation, Edison, NJ, USA